1304.

Preliminary results on PD-1/PD-L1 axis engagement among PD-L1–negative NSCLC patients receiving chemoimmunotherapy

Enrique Caballe-Perez[1,2], Luis Antonio Cabrera-Miranda[1], David Dávila-Dupont[1], Graciela Cruz-Rico[1], Norma Hernández-Pedro[2], Juan Gumuzio[3], Markel Rementeria[3], Jose María Lucio-Lozada[2], Diana Flores-Estrada[1], Cesar Saith Castillo-Ruiz[2], Elí Emmanuel Santana-Aguilar[1] & Oscar Arrieta ✉ [1,2.]

[1] Thoracic Oncology Unit, Instituto Nacional de Cancerología (INCan), Mexico City, México.
[2]Personalized Medicine Laboratory, Instituto Nacional de Cancerología (INCan), Mexico City, México
[3]HAWK Biosystems (FASTBASE Solutions S.L.), Derio, Bizkaia, Spain

Objetives

- To evaluate whether functional quantification of PD-1/PD-L1 engagement predicts clinical outcomes in patients with PD-L1–negative (TPS <1%) NSCLC treated with first-line PD-1/PD-L1–based chemoimmunotherapy.
- To determine if FRET efficiency, as a surrogate for PD-1/PD-L1 complex formation, correlates with progression-free survival (PFS), overall survival (OS), and objective response rate (ORR).

Conclusions

- Quantitative assessment of PD-1/PD-L1 complex formation via FRET efficiency shows promise as a functional biomarker for predicting immunotherapy benefit in patients with PD-L1–negative NSCLC. These findings support further investigation into PD-1/PD-L1 engagement as a predictive tool in this biomarker-limited population.

Plain Language Summary

- **Why did we perform this research?**

Some patients with advanced NSCLC and PD-L1 <1% benefit from chemoimmunotherapy, but we lack reliable biomarkers to predict who will respond.

- **How did we perform this research?**

We used QF-Pro®, an automated FRET-based imaging platform, to measure how strongly PD-1 and PD-L1 interact in tumor samples.

- **What were the findings?**

Patients with high PD-1/PD-L1 engagement had better responses and longer outcomes.

- **What are the implications?**

This approach could help identify PD-L1–negative patients more likely to benefit from immunotherapy.

Scan me!

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials.

Disclosure statement
First and presenting authors have no conflicts of interest to declare.

Corresponding author email address (✉):
ogar@unam.mx

Background

- Chemoimmunotherapy benefits some patients with advanced NSCLC and PD-L1 TPS <1%, but outcomes are heterogeneous and robust predictive biomarkers are lacking [1,2,3]
- Quantifying PD-1/PD-L1 interaction has emerged as a predictive biomarker for patient stratification in anti–PD-1/PD-L1–treated NSCLC and, when combined with PD-L1 expression, may enhance response prediction[4]
- This study aimed to determine whether functional quantification of PD-1/PD-L1 engagement can serve as a predictive biomarker and enable functional stratification of PD-L1–negative NSCLC for anti–PD-1/PD-L1–based chemoimmunotherapy.

Methods

- Retrospective cohort of patients with advanced NSCLC and PD-L1 TPS <1% treated with PD-1/PD-L1–based chemo-immunotherapy; anti–CTLA-4–based regimens were excluded.
- PD-1/PD-L1 interactions were quantified in FFPE tumor samples using QF-Pro®, a high-throughput imaging platform applying quantitative functional proteomics and enhanced Förster resonance energy transfer (FRET); FRET efficiency served as a surrogate for PD-1/PD-L1 complex formation (**Figure 1–2**)
- Patients were stratified by FRET efficiency (highest 40% vs. lower 60%) as determined previously (*Ref. 4*)
- **Primary endpoint:** *Progression-free survival (PFS)* — time from treatment initiation to disease progression or death.
- **Secondary endpoints:**
 - *Overall survival (OS)* — time from treatment initiation to death from any cause.
 - *Objective response rate (ORR)* — proportion of patients achieving CR or PR by RECIST v1.1.
 - *Suboptimal response*: proportion of patients with BOR = SD/PD and PFS <6 mo.

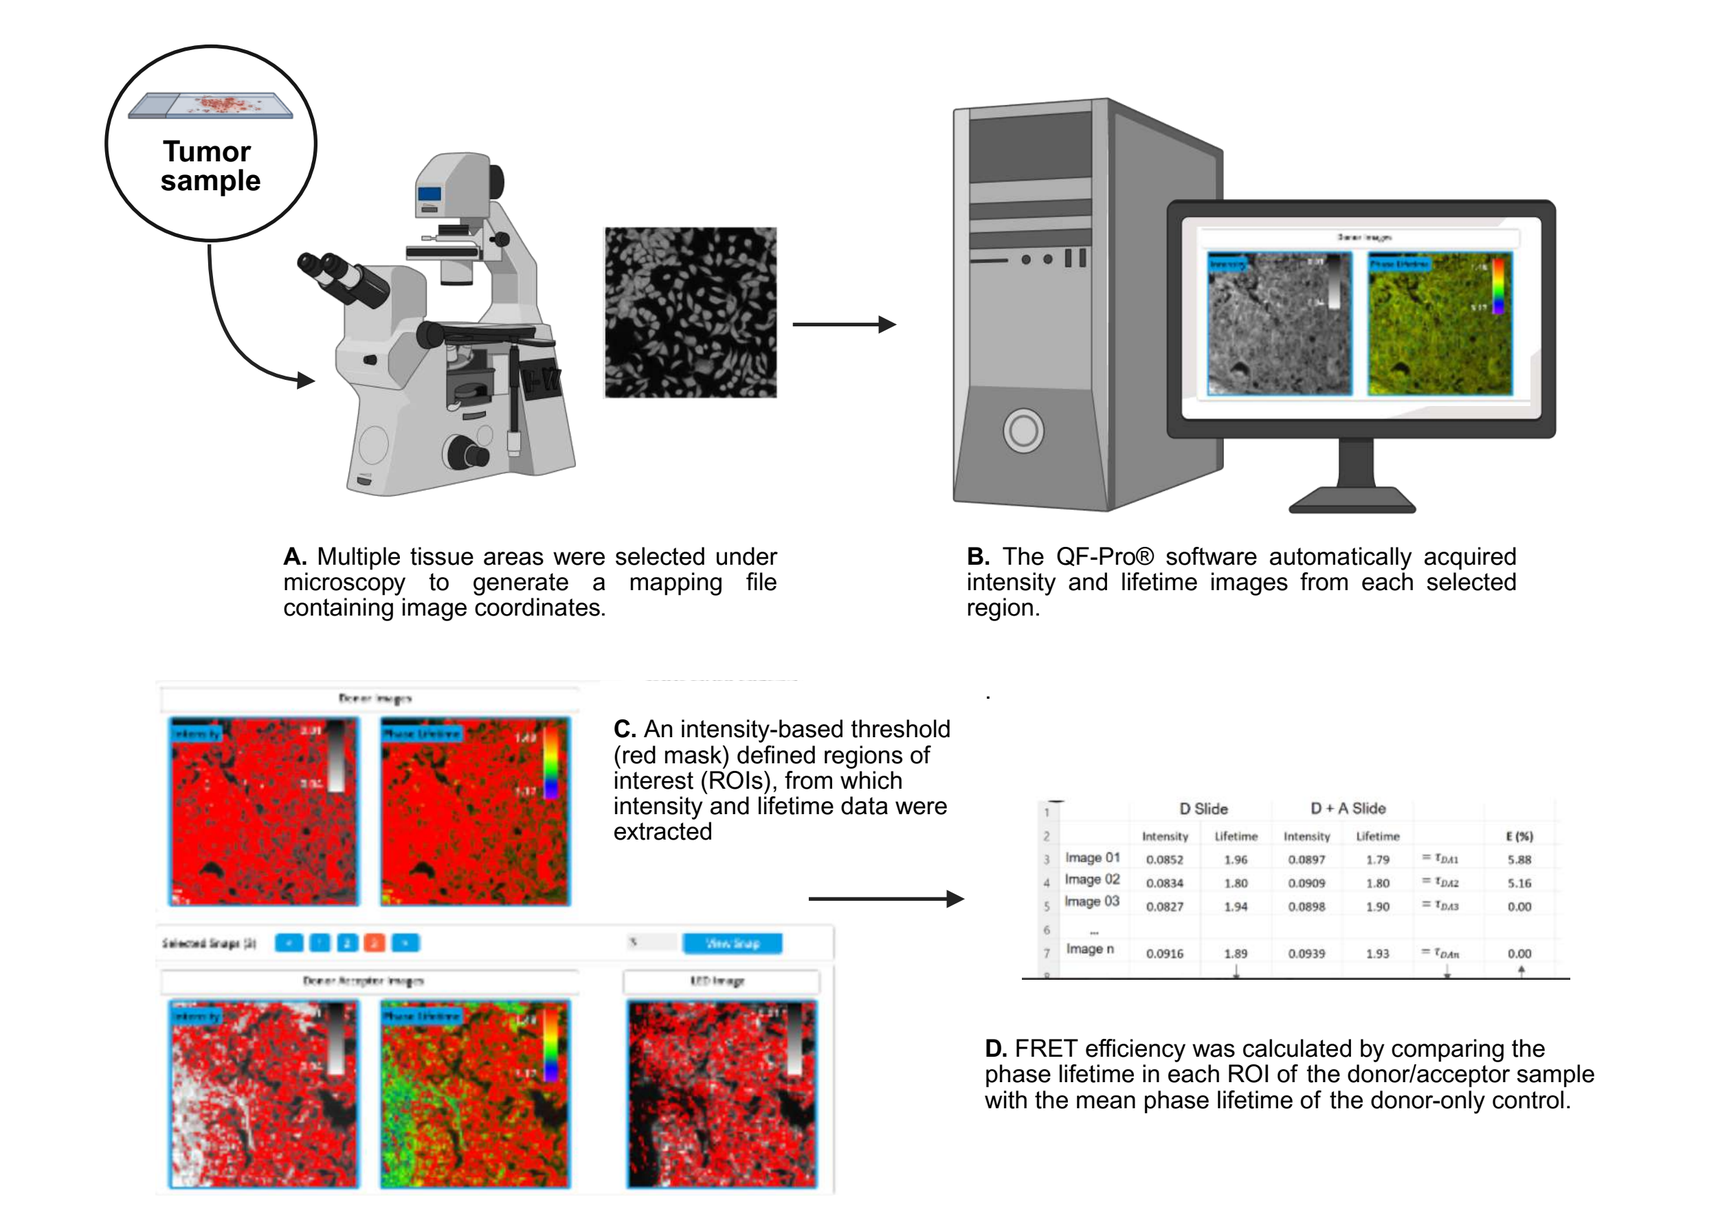
Tumor sample
A. Multiple tissue areas were selected under microscopy to generate a mapping file containing image coordinates.
B. The QF-Pro® software automatically acquired intensity and lifetime images from each selected region.
C. An intensity-based threshold (red mask) defined regions of interest (ROIs), from which intensity and lifetime data were extracted
D. FRET efficiency was calculated by comparing the phase lifetime in each ROI of the donor/acceptor sample with the mean phase lifetime of the donor-only control.

Figure 1. QF-Pro® workflow: A. Tissue regions selected and mapped. **B.** Software auto-acquired intensity and lifetime images. **C.** Intensity threshold (red mask) defined ROIs. **D.** FRET efficiency calculated by comparing phase lifetimes between donor/acceptor and donor-only samples.

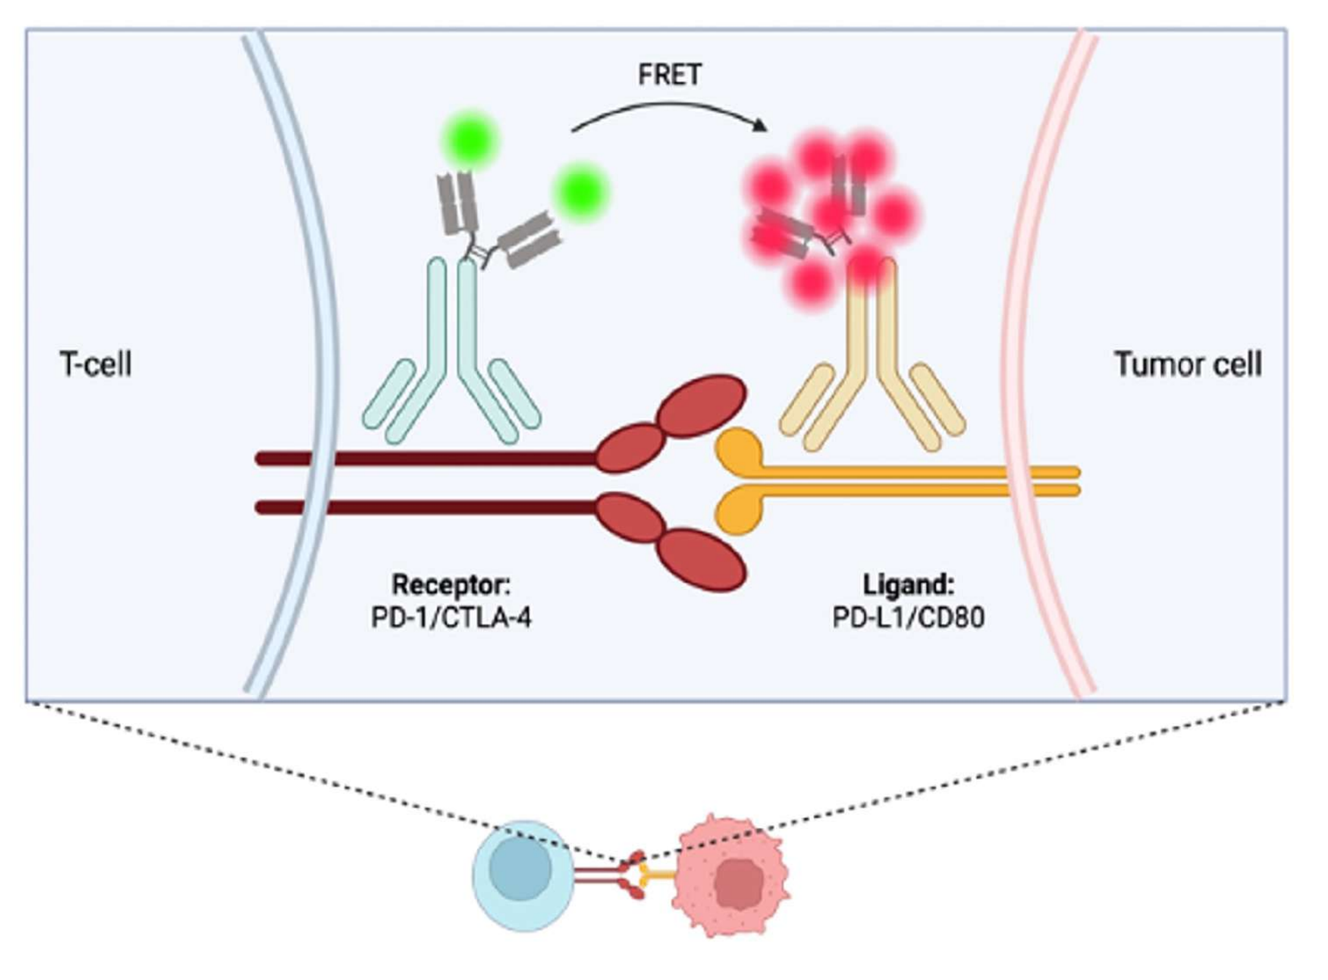
FRET
T-cell
Tumor cell
Receptor: PD-1/CTLA-4
Ligand: PD-L1/CD80

Figure 2. QF-Pro® Assay Scheme. Schematic representation of the QF-Pro® assay for quantifying PD-1/PD-L1 interaction.PD-1 acts as the donor and PD-L1 as the acceptor fluorophore; when both are within <10 nm, measurable FRET energy transfer occurs.

Results

Patient characteristics

- At the data cutoff (June 6, 2025), 27 patients with advanced NSCLC and PD-L1 TPS <1% treated with first-line PD-1/PD-L1–based chemoimmunotherapy were included.
- Patient demographics and clinical characteristics, stratified by FRET efficiency (low ≤60% vs high >40%), are summarized in **Table 1.**

	Mean FRET efficiency (%) Lower 60% FRET	Higher 40% FRET	Total	*P* value
N	15 (55.6)	12 (44.4)	27 (100.0)	
Age, years, median (IQR)	64.5 (60.5 – 68.5)	61.0 (58.0 – 67.0)	63.0 (59.0 – 67.0)	0.590
Sex				
Female	9 (60.0)	4 (33.3)	13 (48.1)	0.168
Male	6 (40.0)	8 (66.7)	14 (51.9)	
ECOG PS, n (%)				
0–1	9 (60.0)	9 (75.0)	18 (66.7)	0.411
≥2	6 (40.0)	3 (25.0)	9 (33.3)	
Smoking History, N (%)				
Positive	10 (66.7)	8 (66.7)	18 (66.7)	1.000
Negative	5 (33.3)	4 (33.3)	9 (33.3)	
Histology, n (%)				
Nonsquamous	13 (86.7)	12 (100.0)	25 (92.6)	0.189
Squamous	2 (13.3)	0 (0.0)	2 (7.4)	
Hepatic metastases, n (%)				
Present	2 (13.3)	3 (25.0)	5 (18.5)	0.438
CNS metastases				
Present	1 (6.7)	6 (50.0)	7 (25.9)	**0.011**
Line of treatment, n (%)				
First line	12 (80.0)	7 (58.3)	19 (70.4)	0.221
Second line	3 (20.0)	5 (41.7)	8 (29.6)	
Anti PD(L)-1 based combo, (%)				
CBDCA/PEM + Durvalumab	2 (13.3)	1 (8.3)	3 (11.1)	0.695
CBDCA/PEM + Pembrolizumab	8 (53.3)	5 (41.7)	13 (48.1)	
CBDCA/PAC + Pembrolizumab	1 (6.7)	1 (8.3)	2 (7.4)	
CBDCA /PAC + Atezolizumab + BEV	1 (6.7)	0 (0.0)	1 (3.7)	
DAC + Pembrolizumab	3 (20.0)	5 (41.7)	8 (29.6)	

Abbreviations: BEV, bevacizumab; CBDCA, carboplatin; DAC, docetaxel; FRET, Förster resonance energy transfer; PEM, pemetrexed; PAC, paclitaxel; ECOG PS, Eastern Cooperative Oncology Group performance status; CNS, central nervous system; IQR, interquartile range. Bold values denote statistical significance at p < 0.05.

- Representative FRET/FLIM images showing high (A) and low (B) PD-1/PD-L1 interaction, with shorter donor lifetimes (red) indicating energy transfer and molecular engagement, are shown in **Figure 3.**

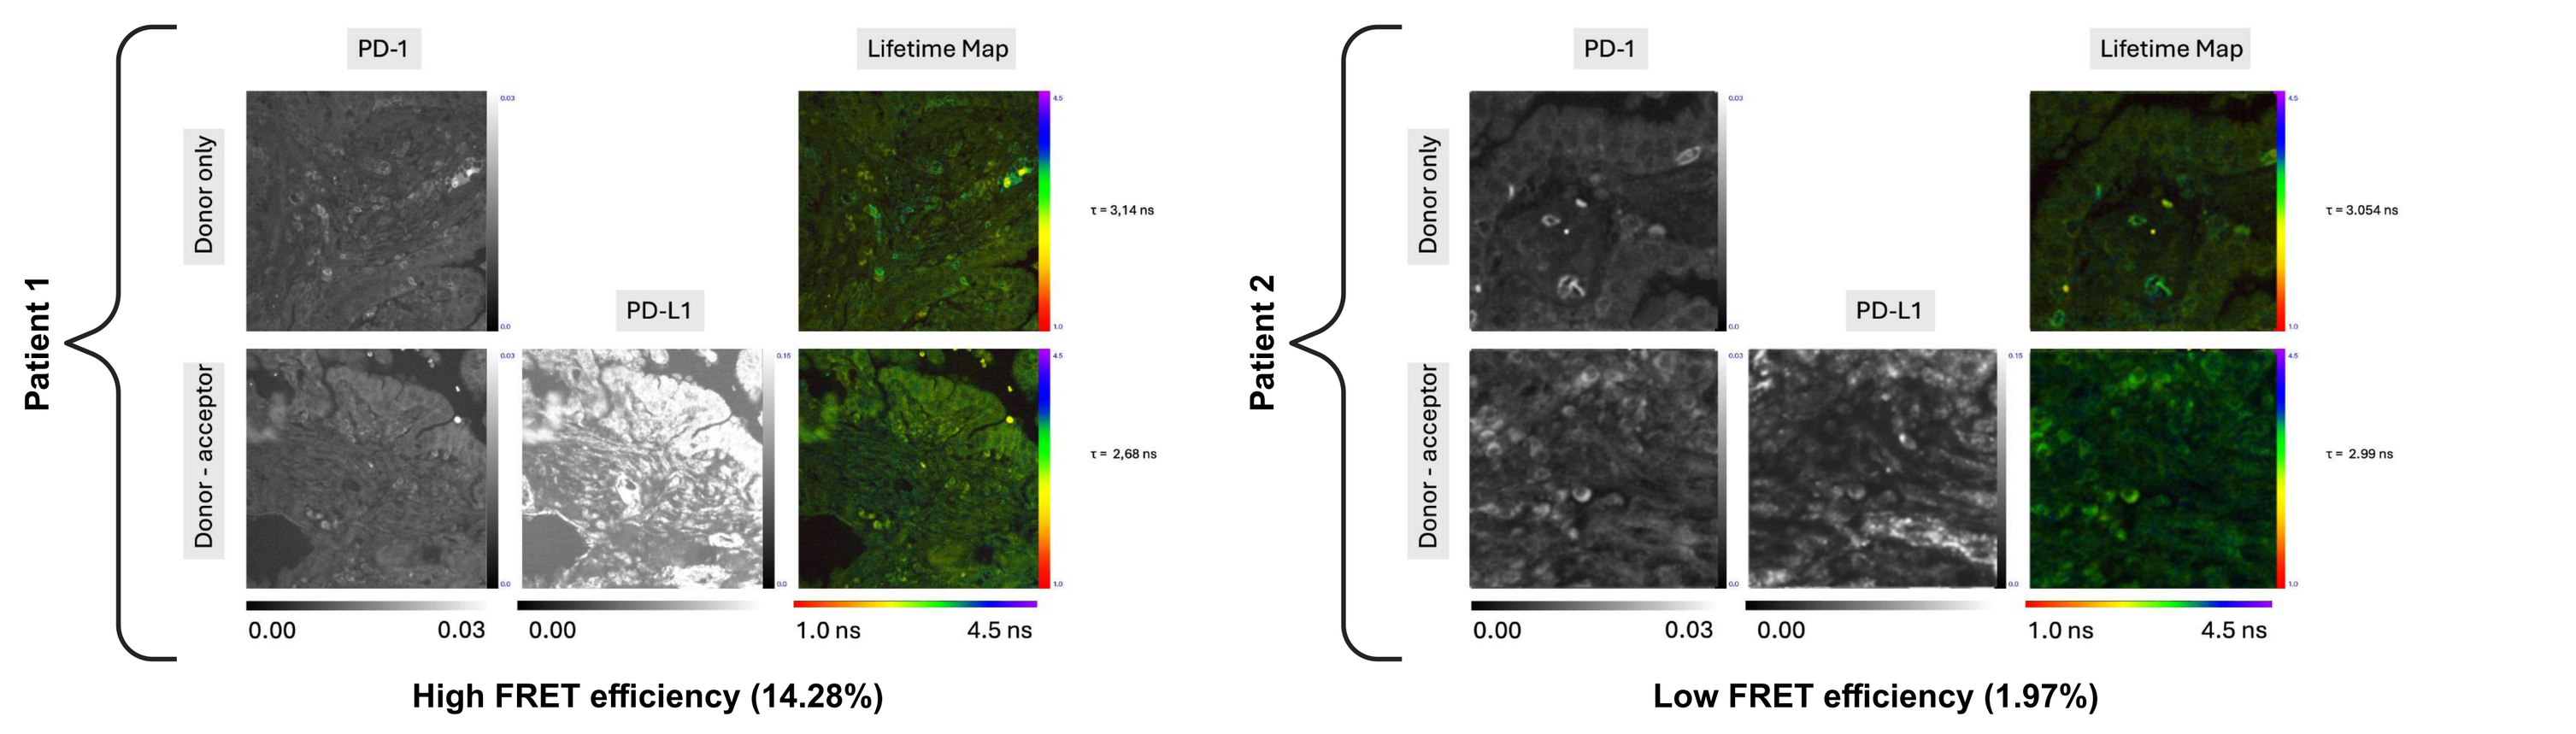
Patient 1
Donor only
Donor - acceptor
PD-1
PD-L1
Lifetime Map
0.00
0.03
1.0 ns
4.5 ns
High FRET efficiency (14.28%)
Patient 2
Low FRET efficiency (1.97%)

Figure 3. Representative FRET/FLIM images of PD-1/PD-L1 interaction (A) High FRET efficiency: Representative images from a patient with high PD-1/PD-L1 interaction, acquired with the FRET/FLIM Violet 3.0 platform. Grayscale panels show PD-1 (donor) and PD-L1 (acceptor) intensity. Pseudo-colour lifetime maps display pixel-wise fluorescence lifetimes (blue = longer ~ 4.5 ns; red = shorter ~ 1 ns). Reduced donor lifetime in the presence of the acceptor indicates energy transfer and thus molecular interaction. **(B) Low FRET efficiency:** Representative images from a patient with low PD-1/PD-L1 interaction acquired under identical conditions. PD-1 and PD-L1 intensity images shown in grayscale; lifetime maps coloured as above. Longer donor lifetimes reflect minimal or absent energy transfer, indicating limited PD-1/PD-L1 engagement.

Association Between PD-1/PD-L1 Engagement and Treatment Response

- The median duration of follow-up was 16.8 months (95% CI, 11.4–22.2)
- Response outcomes in the overall population are shown in **Table 2;** Patients with high FRET efficiency showed a significantly higher ORR compared with those with low FRET efficiency (83.3% vs 33.3%; *p* = 0.009).
- Mean FRET efficiency was significantly higher in patients with an ORR (*p* = 0.042) and lower in those with a suboptimal response (*p* = 0.004), indicating that stronger PD-1/PD-L1 engagement correlates with better treatment outcomes (**Figure 4**)

	Mean FRET efficiency (%) Lower 60% FRET	Higher 40% FRET	Total	*P* value
N	15 (55.6)	12 (44.4)	27 (100.0)	
ORR, % (95% CI)	33.3 (11.8 – 61.6)	83.3 (51.6 – 91.9)	55.6 (35.3 – 74.5)	**0.009**
BOR				
Partial response	5 (33.3)	10 (83.3)	15 (55.6)	**0.017**
Stable disease	6 (40.0)	0 (0.0)	6 (22.2)	
Progressive disease	4 (26.7)	2 (16.7)	6 (22.2)	
Suboptimal response, % (95% CI)	66.7 (38.4 – 88.2)	16.7 (2.1 – 48.4)	44.4 (25.5 – 64.7)	**0.009**

Abbreviations: BOR, best overall response; CI, confidence interval; FRET, Förster resonance energy transfer; ORR, objective response rate. P values derived from Chi-squared test. 95% CIs calculated using the Clopper–Pearson exact method. Bold values denote statistical significance at p < 0.05.

Association Between FRET Efficiency and Survival Outcomes

- mPFS for the overall cohort was 9.2 months (95% CI, 4.0–15.2); patients with high FRET efficiency had a longer PFS than those with low FRET efficiency (14.6 vs 5.4 months; HR 0.42; 95% CI, 0.15–1.14; *p* = 0.083).
- mOS was longer in the high FRET efficiency group compared with the low FRET group (14.8 vs 7.1 months; HR 0.52; 95% CI, 0.18–1.59; *p* = 0.259), although the difference was not statistically significant.
- Patients with higher FRET efficiency showed longer PFS and OS, with a significant positive correlation between FRET efficiency and PFS (r_s = 0.395; *p* = 0.042).

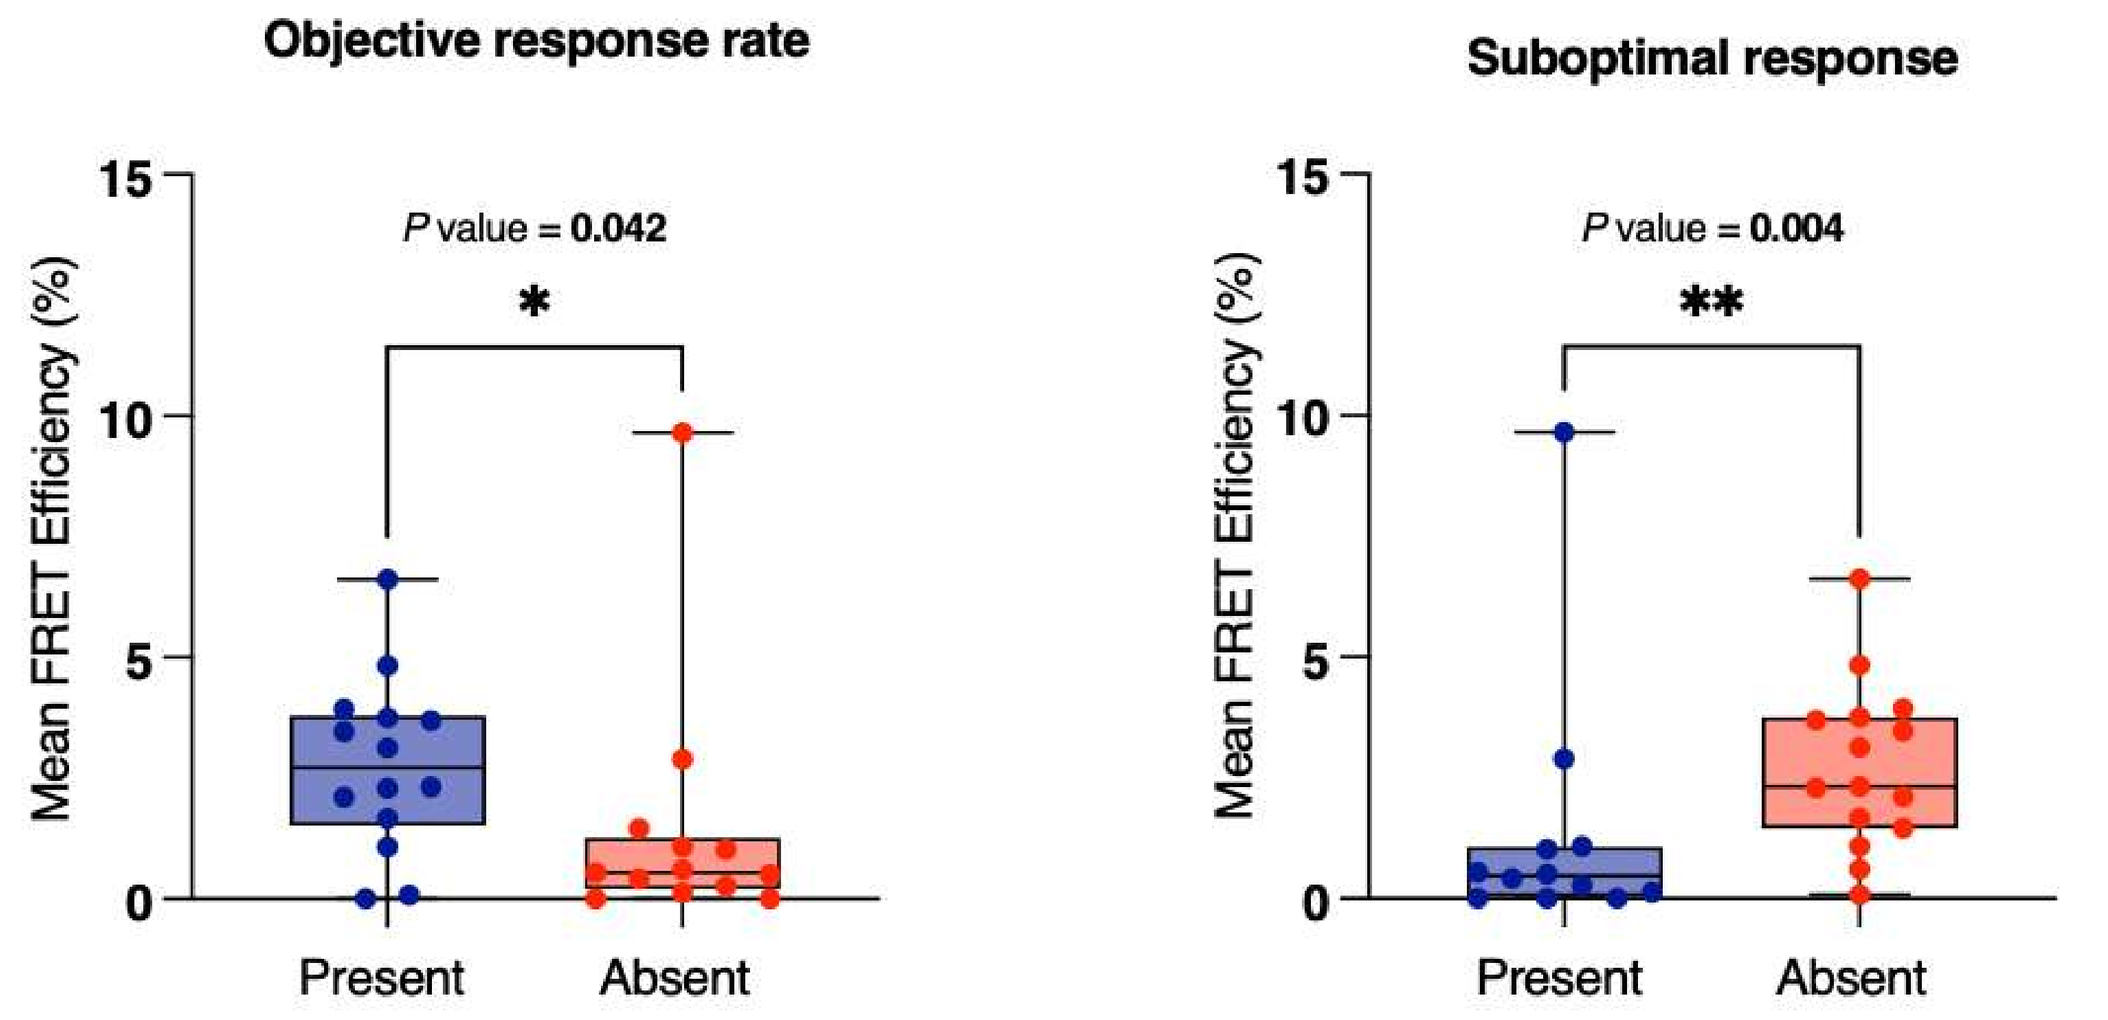
Objective response rate
P value = 0.042
*
Suboptimal response
P value = 0.004
**
Mean FRET Efficiency (%)
Present
Absent

Figure 4. Association between FRET efficiency and treatment response Box plots showing mean FRET efficiency according to objective response rate (left) and suboptimal response (right). Patients with objective response had higher FRET efficiency (*p = 0.042*), while those with suboptimal response showed lower values (*p = 0.004*). *P values calculated using the Mann–Whitney U test; *p < 0.05, ***p < 0.010.*

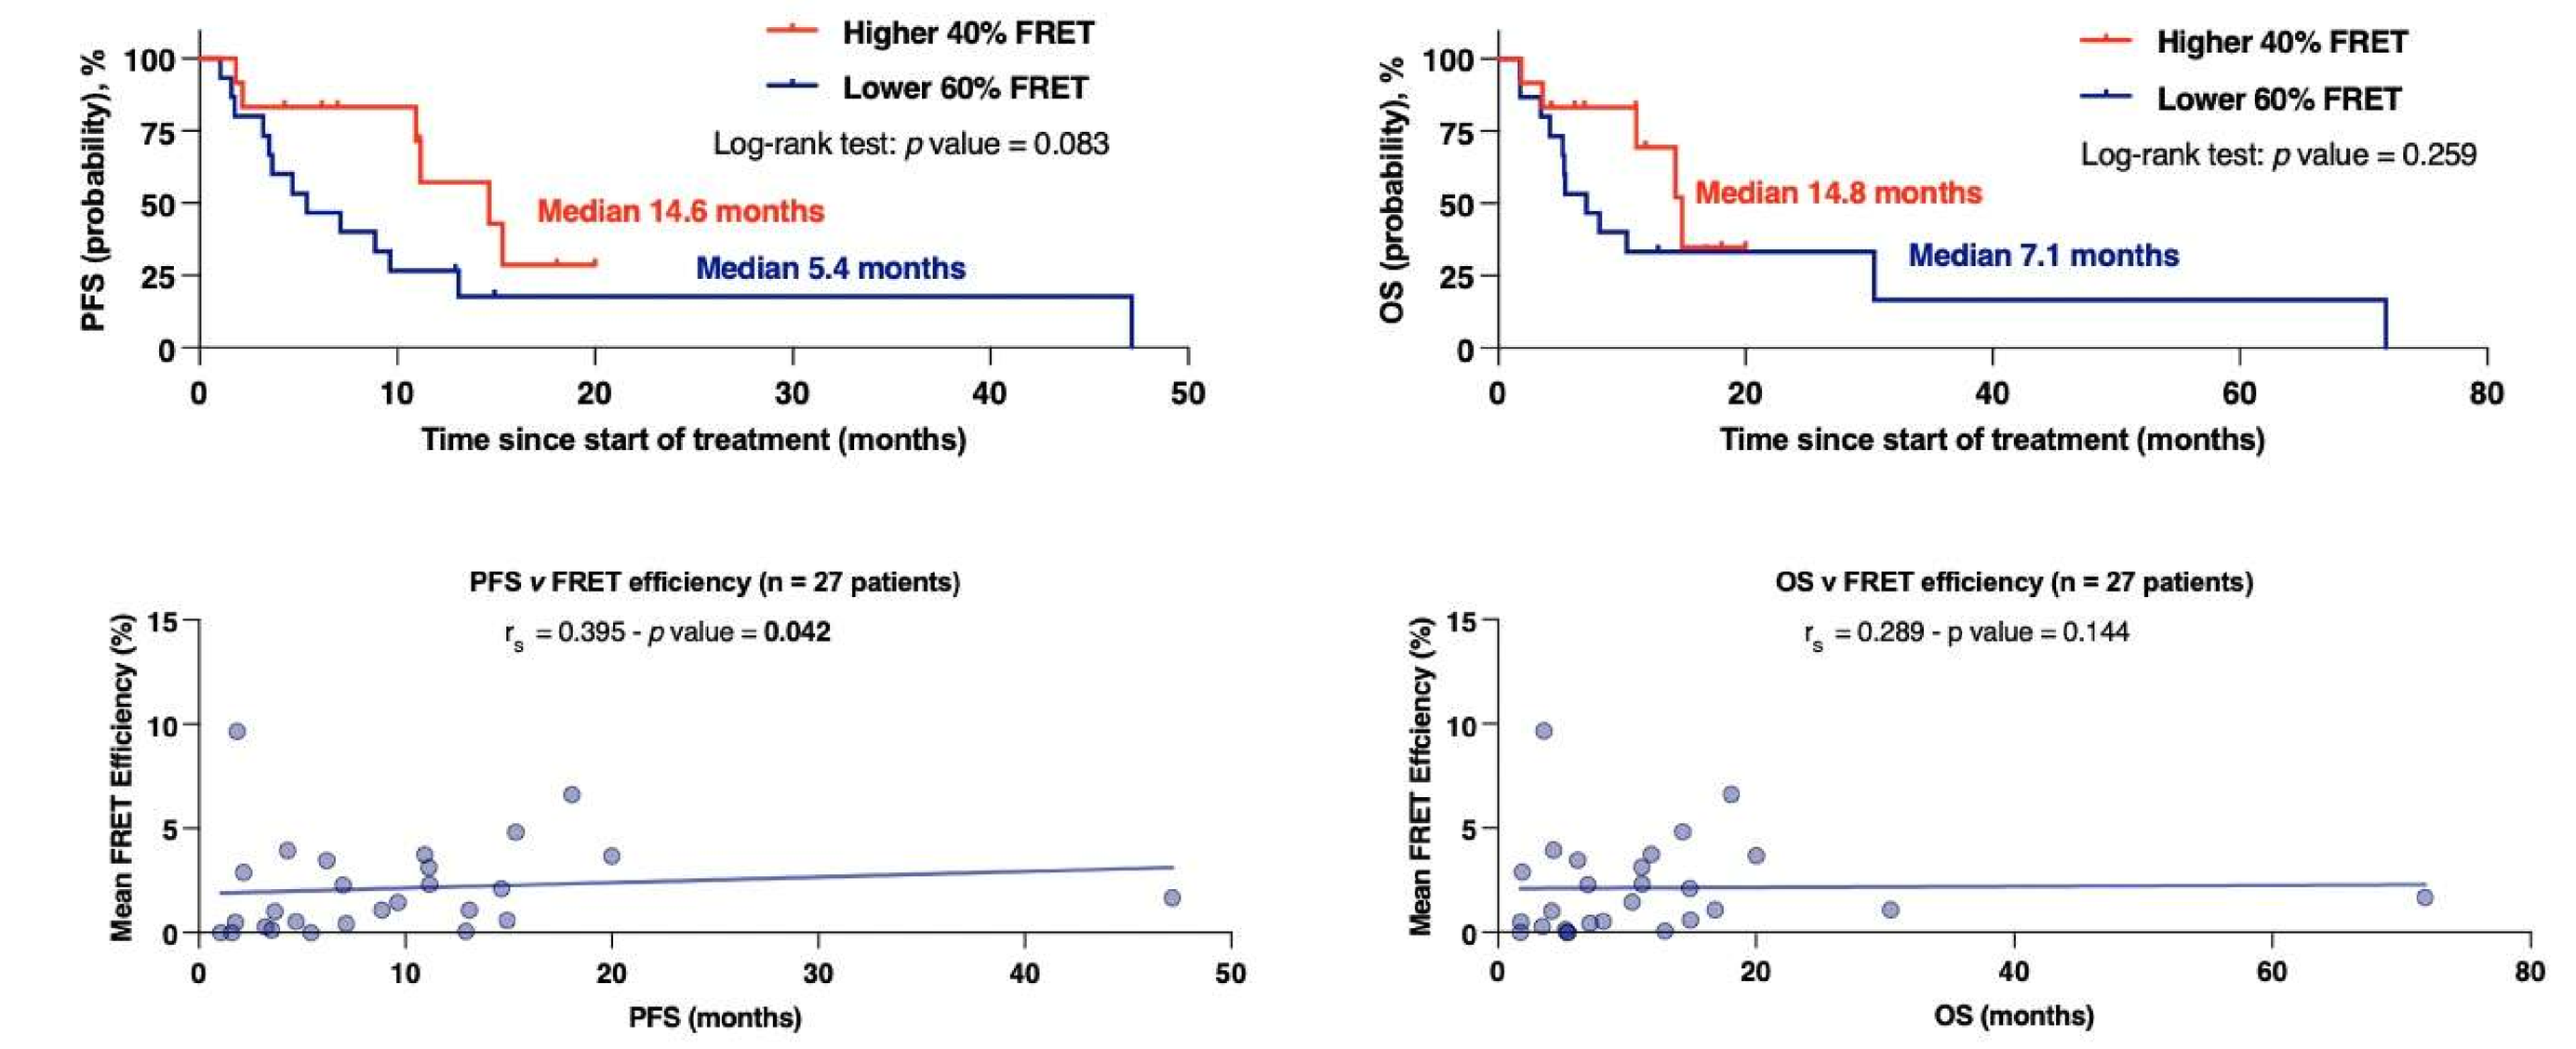
Higher 40% FRET
Lower 60% FRET
Log-rank test: p value = 0.083
Median 14.6 months
Median 5.4 months
PFS (probability), %
Time since start of treatment (months)
Log-rank test: p value = 0.259
Median 14.8 months
Median 7.1 months
OS (probability), %
PFS v FRET efficiency (n = 27 patients)
rs = 0.395 - p value = 0.042
PFS (months)
OS v FRET efficiency (n = 27 patients)
rs = 0.289 - p value = 0.144
OS (months)
Mean FRET Efficiency (%)

Figure 5. Association between FRET efficiency and survival outcomes (A–B) Kaplan–Meier curves for PFS and OS stratified by FRET efficiency (high ≥40% vs low ≤60%). (C–D) Correlation plots showing the relationship between mean FRET efficiency and PFS or OS. Patients with higher FRET efficiency had longer PFS and OS, with a significant correlation between FRET efficiency and PFS (r_s = 0.395; p = 0.042). **Abbreviations**: FRET, Förster resonance energy transfer; PFS, progression-free survival; OS, overall survival. Survival curves compared by the log-rank test; correlations analyzed using Spearman's rank correlation coefficient (r_s).

References

1. Gadgeel SM, Pembrolizumab Plus Chemotherapy for Metastatic NSCLC With Programmed Cell Death Ligand 1 Tumor Proportion Score Less Than 1%: Pooled Analysis of Outcomes After Five Years of Follow-Up. J Thorac Oncol .2024 Aug;19(8):1228-1241. 2. Gemelli M, Efficacy of immune checkpoint inhibitors (ICIs) in PD-L1 negative Non-Small Cell Lung Cancer (NSCLC) - A meta-analysis based on reconstructed individual participant data. Lung Cancer. 2025 Jul;205:108621. 3.Gridelli C, Ardizzoni A, Barberis M, et al: Predictive biomarkers of immunotherapy for non-small cell lung cancer: Results from an experts panel meeting of the Italian association of thoracic oncology. Transl Lung Cancer Res 6:373-386, 2017. 4. Sánchez-Magraner L. Functional Engagement of the PD-1/PD-L1 Complex But Not PD-L1 Expression Is Highly Predictive of Patient Response to Immunotherapy in Non-Small-Cell Lung Cancer J Clin Oncol. 2023 May 10;41(14):2561-2570

Acknowledgements

Áviles-Salas Alejandro, Pathology Unit, Instituto Nacional de Cancerología
Madrid Norma, HAWK Biosystems

HAWK Biosystems | Salud Secretaría de Salud | Instituto Nacional de Cancerología

Created with BioRender Poster Builder